Latest findings from the breast cancer cohort in SUMMIT – a phase 2 ‘basket’ trial of neratinib + trastuzumab + fulvestrant for *HER2*-mutant, hormone receptor-positive, metastatic breast cancer

Komal Jhaveri,[1] Cristina Saura,[2] Angel Guerrero-Zotano,[3] Iben Spanggaard,[4] François-Clement Bidard,[5] Jonathan W Goldman,[6] José A García-Sáenz,[7] Andrés Cervantes,[8] Valentina Boni,[9] John Crown,[10] Adam Brufsky,[11] Sherene Loi,[12] Barbara Haley,[13] Ingrid A Mayer,[14] Stephen Chia,[15] Janice Lu,[16] James Waisman,[17] Noa Efrat Ben-Baruch,[18] Mark E Burkard,[19] Jennifer M Suga,[20] Lucía González-Cortijo,[21] Bruno Perruccі,[22] Feng Xu,[23] Sofia Wong,[23] Jie Zhang,[23] Lisa D Eli,[23] Alshad S Lalani,[23] and Hans Wildiers[24]

[1]Memorial Sloan Kettering Cancer Center, New York, NY; [2]Vall d'Hebron University Hospital, Vall d'Hebron Institute of Oncology, Barcelona, Spain; [3]Fundación Instituto Valenciano de Oncología, Valencia, Spain; [4]Rigshospitalet, Copenhagen University Hospital, Copenhagen, Denmark; [5]Institut Curie, St Cloud, France; [6]UCLA, Santa Monica, CA; [7]Hospital Clínico San Carlos, Madrid, Spain; [8]Hospital Clínico Universitario, University of Valencia, Valencia, Spain; [9]START Madrid-CIOCC, Hospital Universitario, Madrid Sanchinarro, Madrid, Spain; [10]St. Vincent's University Hospital, Dublin, Ireland; [11]Magee-Womens Hospital of UPMC, Pittsburgh, PA; [12]Peter MacCallum Cancer Centre, Melbourne, Australia; [13]UT Southwestern Medical Center, Dallas, TX; [14]Vanderbilt University Medical Center/Vanderbilt-Ingram Cancer Center, Nashville, TN; [15]British Columbia Cancer Agency, University of British Columbia, Vancouver, BC, Canada; [16]USC Norris Comprehensive Cancer Center, Los Angeles, CA; [17]City of Hope Comprehensive Cancer Center, Duarte, CA; [18]Kaplan Medical Center, Rehovot, Israel; [19]University of Wisconsin School of Medicine and Public Health, Madison, WI; [20]Kaiser Permanente Cancer Research Center, Vallejo, CA; [21]Hospital Universitario Quironsalud, Madrid, Spain; [22]ASST Cremona, Cremona, Italy; [23]Puma Biotechnology Inc., South San Francisco, CA; [24]University Hospitals Leuven, Leuven, Belgium

#PD1-05

Introduction

- *HER2* mutations occur in approximately 2% of primary breast cancers and in 7–8% of hormone receptor positive (HR+) metastatic breast cancer (MBC) and have a unique mechanism of oncogenic addiction to HER2 signaling:[1–3]
 - Acquired HER2 mutations may confer resistance to endocrine-based therapies.[3,4]
- Neratinib is an oral, irreversible, pan-HER tyrosine kinase inhibitor that has demonstrated encouraging clinical activity either as a single agent or in combination with fulvestrant in *HER2*-mutated, HER2-non-amplified MBC in the SUMMIT basket trial.[5–7]
- ctDNA analysis of *HER2*-mutated patients from SUMMIT that benefited from neratinib as a single agent or in combination with fulvestrant revealed acquisition of secondary *HER2* mutations and/or *HER2* gene amplification upon progression:[6]
 - Suggests that the combination of neratinib + trastuzumab may improve durability of responses.

Objective

- We investigated whether addition of trastuzumab to neratinib + fulvestrant could further improve clinical benefit in a cohort of patients with *HER2*-mutant, HR+ MBC from SUMMIT.

Methods

Figure 1. Current SUMMIT study design

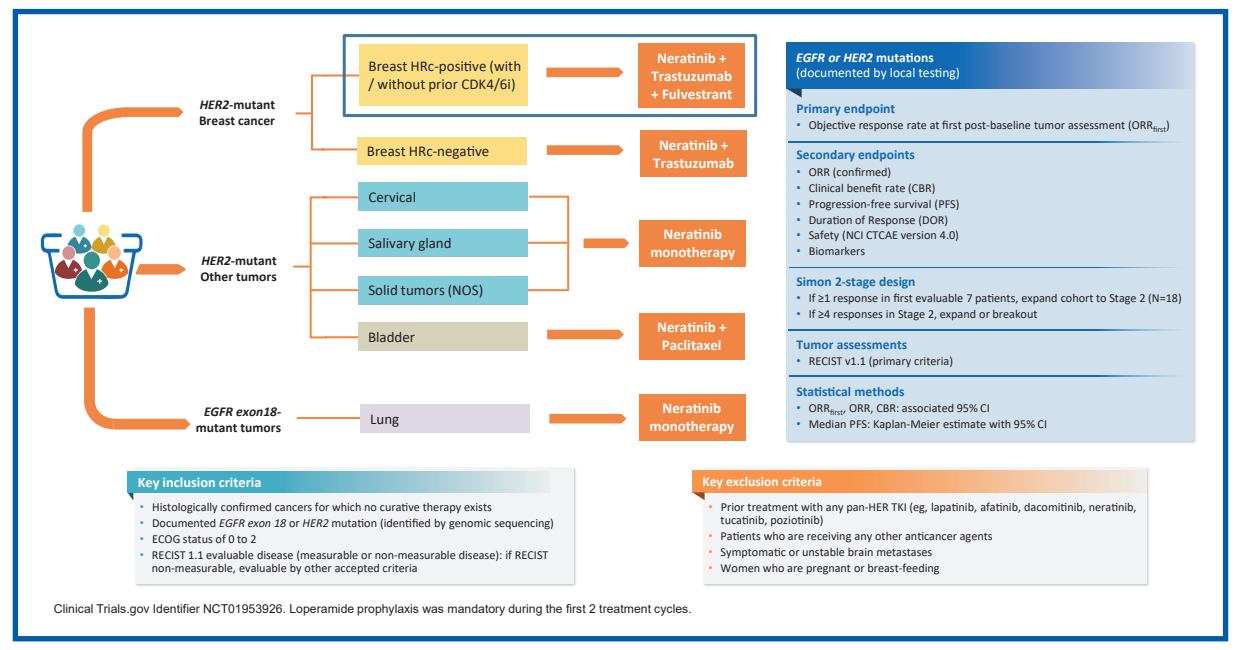
HER2-mutant Breast cancer
Breast HRc-positive (with / without prior CDK4/6i)
Neratinib + Trastuzumab + Fulvestrant
Breast HRc-negative
Neratinib + Trastuzumab
HER2-mutant Other tumors
Cervical
Salivary gland
Solid tumors (NOS)
Neratinib monotherapy
Bladder
Neratinib + Paclitaxel
EGFR exon18-mutant tumors
Lung
Neratinib monotherapy
EGFR or HER2 mutations (documented by local testing)
Primary endpoint
Objective response rate at first post-baseline tumor assessment (ORRfirst)
Secondary endpoints
ORR (confirmed)
Clinical benefit rate (CBR)
Progression-free survival (PFS)
Duration of Response (DOR)
Safety (NCI CTCAE version 4.0)
Biomarkers
Simon 2-stage design
If ≥1 response in first evaluable 7 patients, expand cohort to Stage 2 (N=18)
If ≥4 responses in Stage 2, expand or breakout
Tumor assessments
RECIST v1.1 (primary criteria)
Statistical methods
ORRfirst, ORR, CBR: associated 95% CI
Median PFS: Kaplan-Meier estimate with 95% CI
Key inclusion criteria
Histologically confirmed cancers for which no curative therapy exists
Documented EGFR exon 18 or HER2 mutation (identified by genomic sequencing)
ECOG status of 0 to 2
RECIST 1.1 evaluable disease (measurable or non-measurable disease): if RECIST non-measurable, evaluable by other accepted criteria
Key exclusion criteria
Prior treatment with any pan-HER TKI (eg, lapatinib, afatinib, dacomitinib, neratinib, tucatinib, poziotinib)
Patients who are receiving any other anticancer agents
Symptomatic or unstable brain metastases
Women who are pregnant or breast-feeding

Clinical Trials.gov Identifier NCT01953926. Loperamide prophylaxis was mandatory during the first 2 treatment cycles.

Results

Table 1. Baseline demographics

Patient characteristics	Safety evaluable patients (n=51)
Median (range), years	58 (25–82)
<65 years, n (%)	36 (70.6)
≥65 years, n (%)	15 (29.4)
Gender, n (%)	
Female	51 (100)
ECOG performance status, n (%)	
0	24 (47.1)
1	26 (51.0)
2	1 (2.0)
Menopausal status, n (%)	
Post-menopausal	45 (88.2)
Pre-menopausal	6 (11.8)
Disease characteristics	
Histological type, n (%)	
Ductal	16 (31.4)
Lobular	33 (64.7)
Mixed ductal and lobular	1 (2.0)
Other	1 (2.0)
HER2 status[a], n (%)	
Negative	49 (96.1)
Equivocal	2 (3.9)
HR (ER/PR) status, n (%)	
HR+ (ER+ and/or PR+)	51 (100)
Location of disease at time of enrollment, n (%)	
Visceral	43 (84.3)
Non-visceral only	8 (15.7)
Median time from first metastasis to enrollment, years (range)	2.90 (0.2–9.1)

Safety evaluable: all enrolled patients who received at least 1 dose of neratinib.
ECOG: Eastern Cooperative Oncology Group.
[a]Negative: IHC=0 or 1+; or FISH (ISH) HER2/CEP 17 ratio <2.0; or FISH (ISH) HER2 gene copy # <4.0. Equivocal: IHC=2+; or FISH (ISH) HER2 gene copy # ≥4.0 and <6.0. Unavailable: data entry pending.

Table 2. Prior therapies in the locally advanced/metastatic setting

Prior therapies	Safety evaluable patients (n=51)
Patients with prior treatment for locally advanced/metastatic disease, n (%)	46 (90.2)[a]
Median number of prior therapies (range)	4 (1–10)
Prior endocrine therapy, n (%)	
Prior aromatase inhibitor	35 (68.6)
Prior fulvestrant	36 (70.6)
Prior tamoxifen	4 (7.8)
Prior chemotherapy, n (%)	35 (68.6)
Prior HER2 antibody-directed therapy, n (%)	2 (3.9)[a]
Prior CDK4/6 inhibitor, n (%)	30 (58.8)
Prior PIK3CA inhibitor, n (%)	4 (7.9)
Prior mTOR inhibitor, n (%)	15 (29.4)

[a]Two patients received prior treatment of trastuzumab + pertuzumab + docetaxel. Five patients did not receive prior treatments for metastatic disease: for one patient, the data was entered after the data snapshot for this poster and this patient had four prior lines of therapy for metastatic disease; for the other four patients no prior therapy for metastatic disease was recorded.

Table 3. Subject disposition

Parameter	Safety evaluable patients (n=51)
Median duration of treatment, months (range)	6.7 (0.9–31.6)
Patients continuing treatment, n (%)	18 (35.3)
Treatment discontinuation, n (%)	33 (64.7)
Disease progression	30 (58.8)
Death	0
Adverse event	1 (2.0)
Other[a]	2 (4.0)

[a]One patient discontinued the treatment due to clinical progression and the other patient due to the treating physician's decision.

Table 4. Efficacy summary (RECIST evaluable patients, n=37)

Parameter	RECIST evaluable patients (n=37)	Subgroups: Prior CDK4/6i (n=23)	Subgroups: Prior fulvestrant (n=25)
Objective response (confirmed)[a] n (%)	17 (45.9)	9 (39.1)	11 (44.0)
CR	1 (2.7)	0	0
PR	16 (43.2)	9 (39.1)	11 (44.0)
Objective response rate, % (95% CI)	**45.9** (29.5–63.1)	**39.1** (19.7–61.5)	**44.0** (24.4–65.1)
Best overall response, n (%)	21 (56.8)	11 (47.8)	13 (52.0)
CR	1 (2.7)	0	0
PR	20 (54.1)	11 (47.8)	13 (52.0)
Best overall response rate, % (95% CI)	**56.8** (39.5–72.9)	**47.8** (26.8–69.4)	**52.0** (31.3–72.2)
Median[b] DOR, months (95% CI)	**10.9** (6.4–NE)	**8.7** (6.4–10.9)	**8.4** (5.8–12.5)
Clinical benefit[c] n (%)	20 (54.1)	12 (52.2)	14 (56.0)
CR or PR	17 (45.9)	9 (39.1)	11 (44.0)
SD ≥24 weeks	3 (8.1)	3 (13.0)	3 (12.0)
Clinical benefit rate, % (95% CI)	**54.1** (36.9–70.5)	**52.2** (30.6–73.2)	**56.0** (34.9–75.6)
Median[b] PFS time to event, months (95% CI)	**8.3** (4.2–14.5)	**8.2** (4.0–15.1)	**8.3** (3.1–12.5)

Data cut-off: 16 October 2020. DOR, duration of response; NE, not estimable; PFS, progression-free survival. RECIST evaluable: patients with RECIST measurable disease at baseline with at least 1 post-baseline tumor assessment.
[a]Objective response rate (ORR) is defined as either a complete or partial response that is confirmed no less than 4-weeks after the criteria for response are initially met; bKaplan-Meier analysis; cClinical benefit rate (CBR) is defined as confirmed CR or PR or stable disease (SD) for ≥24 weeks (within +/– 7-day visit window).

Figure 2. Distribution of *HER2* mutations (RECIST evaluable patients, n=35)[a]

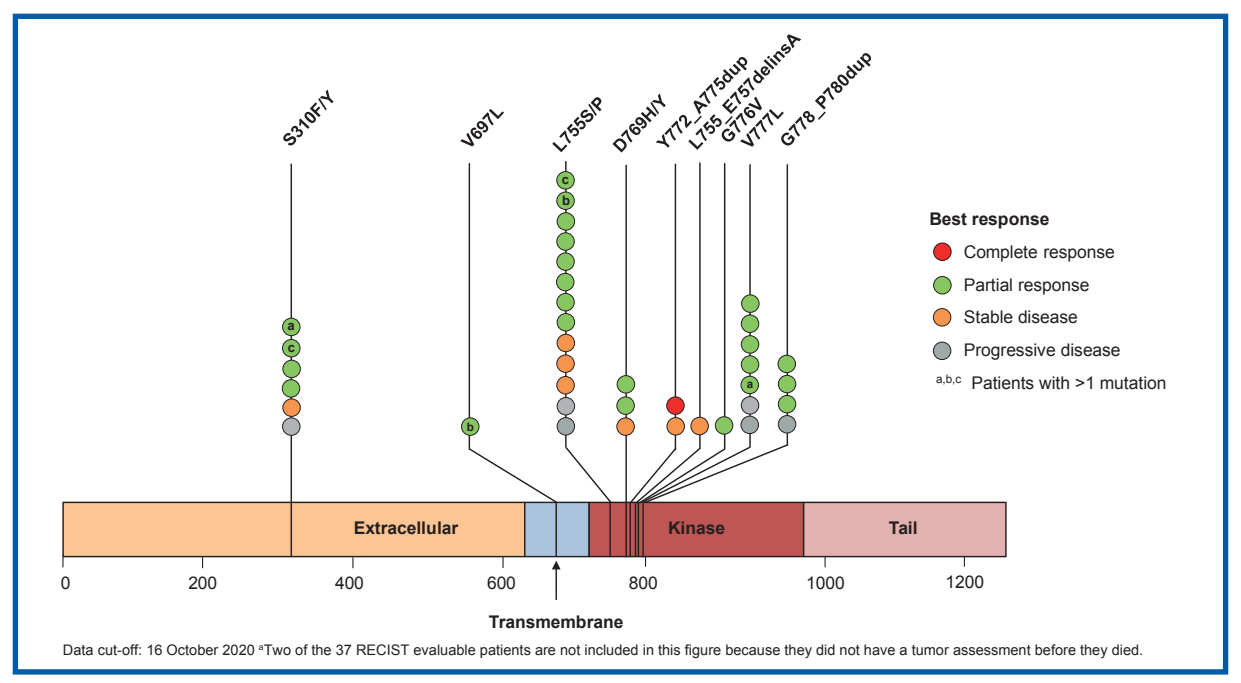
S310F/Y
V697L
L755S/P
D769H/Y
Y772_A775dup
L755_E757delinsA
G776V
V777L
G778_P780dup
Best response
Complete response
Partial response
Stable disease
Progressive disease
a,b,c Patients with >1 mutation
Extracellular
Kinase
Tail
0
200
400
600
800
1000
1200
Transmembrane

Data cut-off: 16 October 2020 [a]Two of the 37 RECIST evaluable patients are not included in this figure because they did not have a tumor assessment before they died.

Figure 3. Change in tumor size and characteristics (n=35)[a]

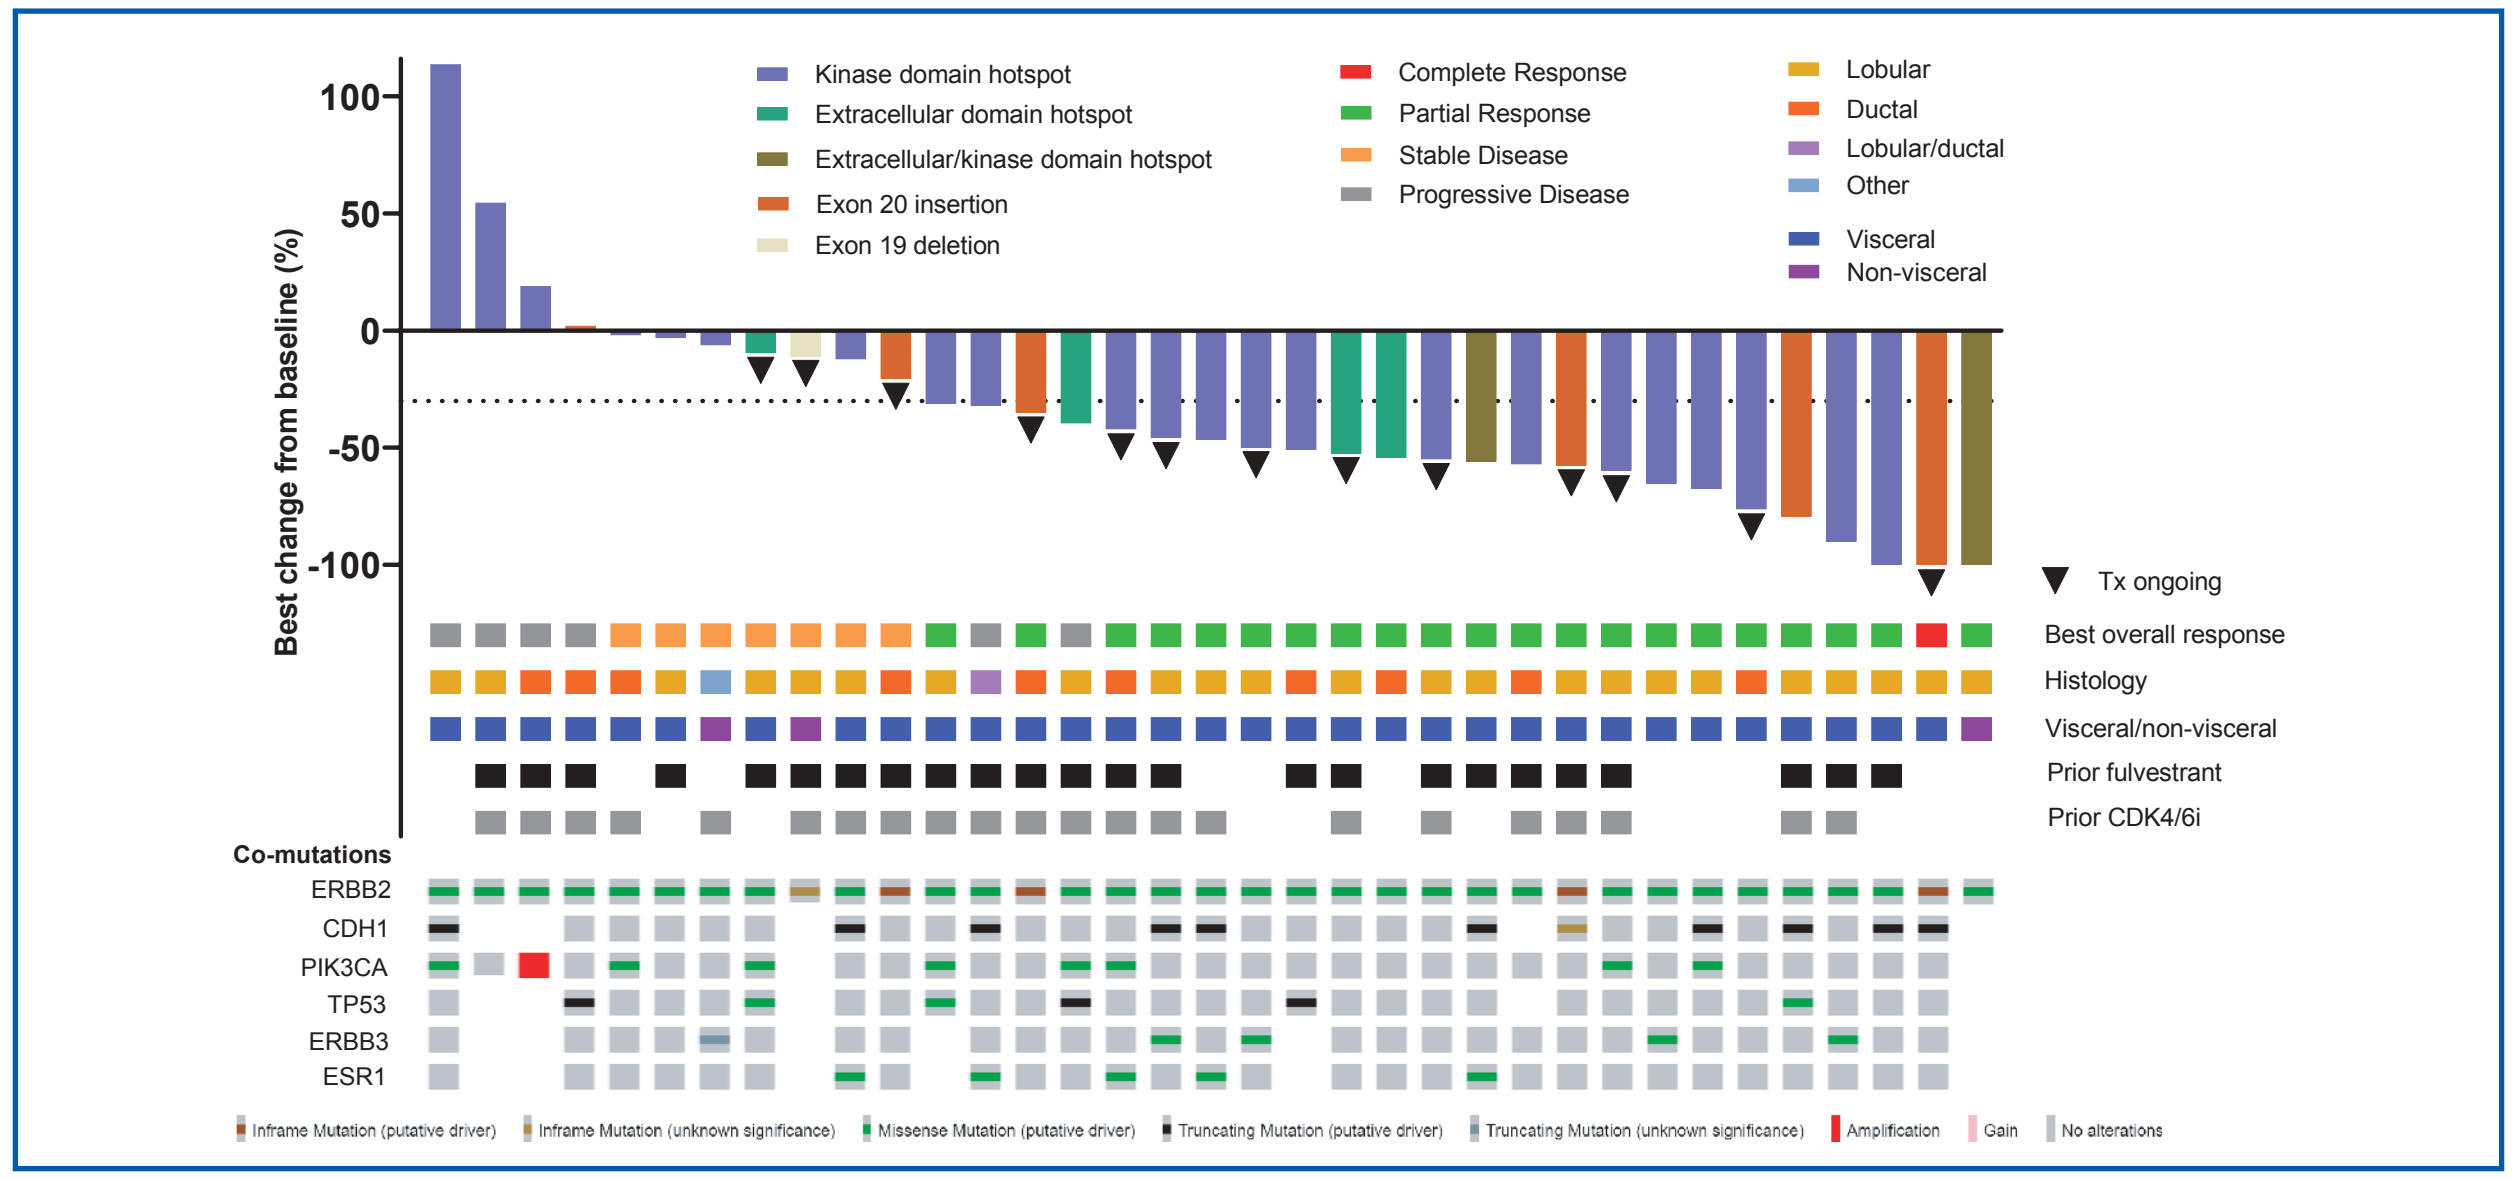
100
50
0
-50
-100
Best change from baseline (%)
Kinase domain hotspot
Extracellular domain hotspot
Extracellular/kinase domain hotspot
Exon 20 insertion
Exon 19 deletion
Complete Response
Partial Response
Stable Disease
Progressive Disease
Lobular
Ductal
Lobular/ductal
Other
Visceral
Non-visceral
Tx ongoing
Best overall response
Histology
Visceral/non-visceral
Prior fulvestrant
Prior CDK4/6i
Co-mutations
ERBB2
CDH1
PIK3CA
TP53
ERBB3
ESR1
Inframe Mutation (putative driver)
Inframe Mutation (unknown significance)
Missense Mutation (putative driver)
Truncating Mutation (putative driver)
Truncating Mutation (unknown significance)
Amplification
Gain
No alterations

Data cut-off: 16 October 2020.
[a]Two of the 37 RECIST evaluable patients are not included in this figure because they did not have a tumor assessment before they died.

Figure 4. Duration of treatment and best response (RECIST evaluable patients, n=37)

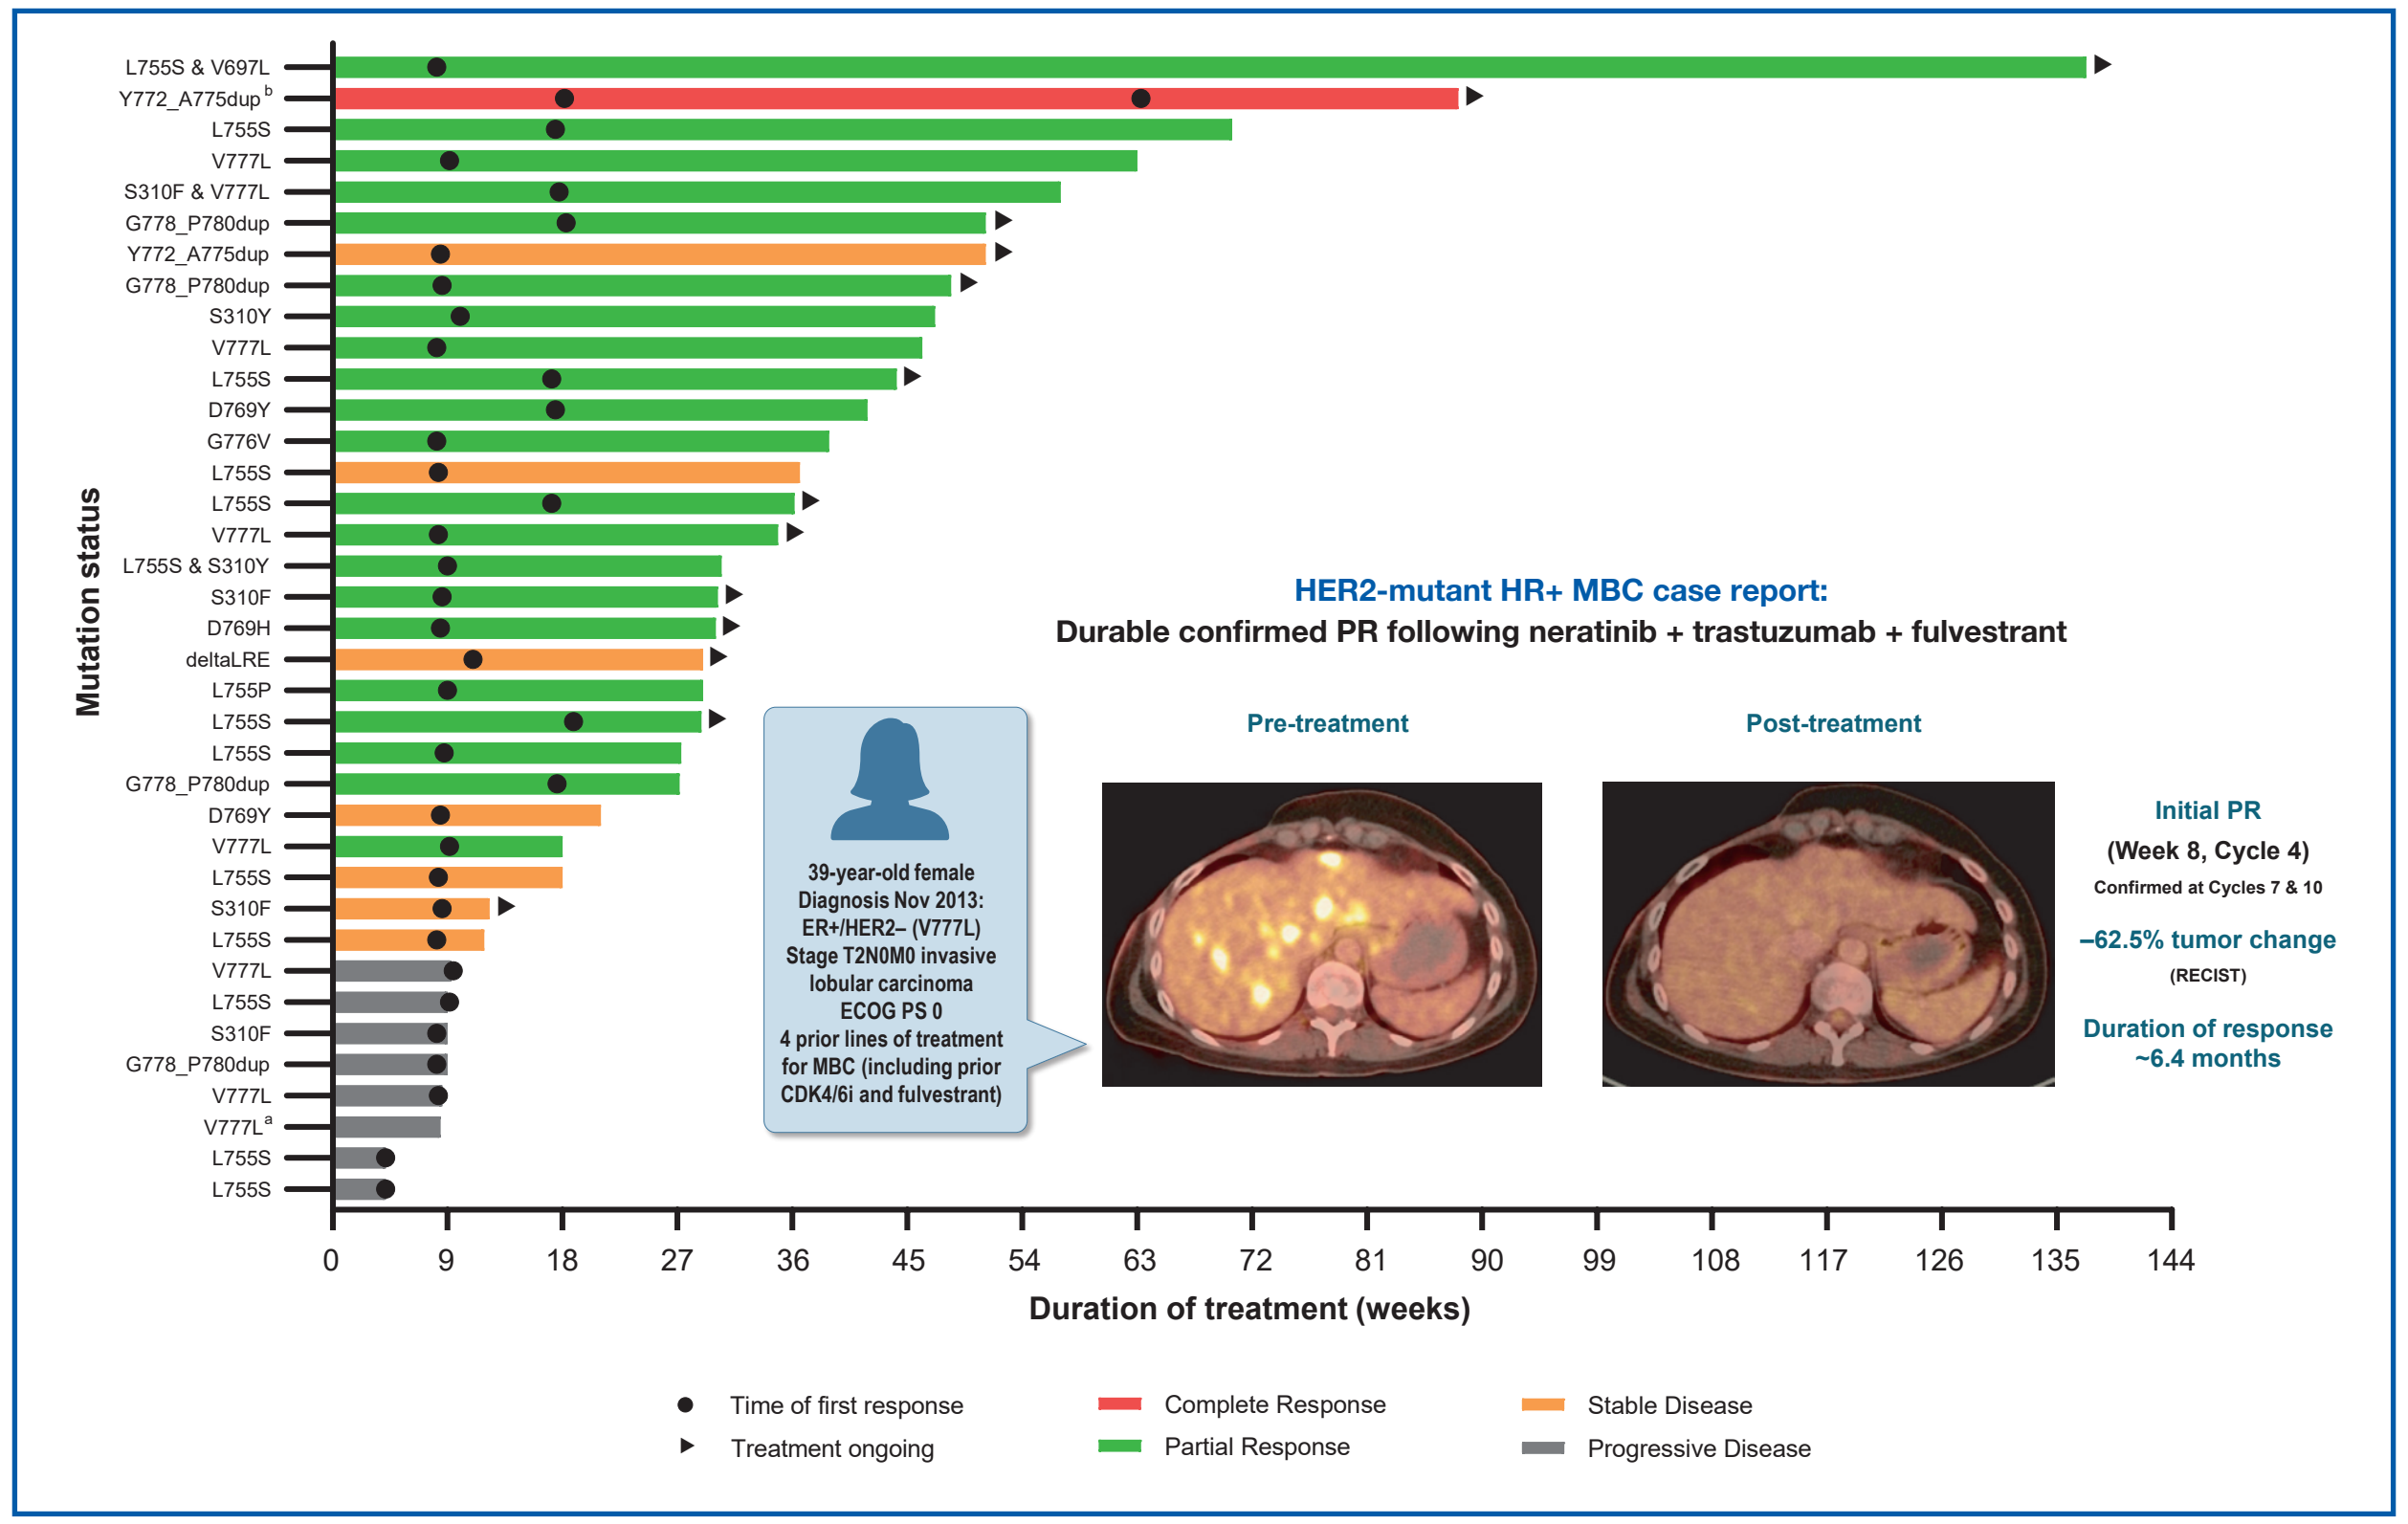
L755S & V697L
Y772_A775dup b
L755S
V777L
S310F & V777L
G778_P780dup
Y772_A775dup
G778_P780dup
S310Y
V777L
L755S
D769Y
G776V
L755S
L755S
V777L
L755S & S310Y
S310F
D769H
deltaLRE
L755P
L755S
L755S
G778_P780dup
D769Y
V777L
L755S
S310F
L755S
V777L
L755S
S310F
G778_P780dup
V777L
V777L a
L755S
L755S
Mutation status
0
9
18
27
36
45
54
63
72
81
90
99
108
117
126
135
144
Duration of treatment (weeks)
Time of first response
Treatment ongoing
Complete Response
Partial Response
Stable Disease
Progressive Disease
HER2-mutant HR+ MBC case report:
Durable confirmed PR following neratinib + trastuzumab + fulvestrant
Pre-treatment
Post-treatment
39-year-old female
Diagnosis Nov 2013:
ER+/HER2– (V777L)
Stage T2N0M0 invasive lobular carcinoma
ECOG PS 0
4 prior lines of treatment for MBC (including prior CDK4/6i and fulvestrant)
Initial PR
(Week 8, Cycle 4)
Confirmed at Cycles 7 & 10
–62.5% tumor change
(RECIST)
Duration of response
~6.4 months

Data cut-off: 16 October 2020.
[a]This patient died in hospice due to clinical progression and did not have a tumor assessment before she died.
[b]This patient had a first partial response at week 18 and a first complete response at week 63.

Table 5. Most common treatment-emergent adverse events

Adverse event, n (%)	Safety evaluable patients (n=51): All grade	Safety evaluable patients (n=51): Grade 3 or 4
Subjects with at least 1 adverse event, n (%)	49 (96.1)	33 (64.7)
Diarrhea	45 (88.2)	20 (39.2)[a]
Nausea	34 (66.7)	0
Constipation	21 (41.2)	0
Fatigue	18 (35.3)	3 (5.9)
Vomiting	22 (43.1)	2 (3.9)
Decreased appetite	20 (39.2)	3 (5.9)
Abdominal pain	12 (23.5)	0
Headache	8 (15.7)	0

[a]No Grade 4 diarrhea was reported.

Table 6. Characteristics of diarrhea

	Safety evaluable patients (n=51)
Incidence of diarrhea, n (%)[a]	
Any grade	45 (88.2)
Grade 1	13 (25.5)
Grade 2	12 (23.5)
Grade 3	20 (39.2)
Action taken with neratinib, n (%)	
Leading to temporary hold	21 (41.2)
Leading to dose reduction	11 (21.6)
Leading to permanent discontinuation	0
Leading to hospitalization	1 (2.0)
Median cumulative duration of grade 3 diarrhea per patient (Q1, Q3), days	6 (1–16.5)

Conclusions

- *HER2* mutations are oncogenic in a subset of MBC and are clinically actionable with HER2-directed therapies.
- The combination of neratinib + fulvestrant + trastuzumab demonstrated encouraging clinical activity in heavily pre-treated *HER2*-mutant, HR+, HER2-non-amplified MBC, including patients who had previously received either fulvestrant and/or CDK4/6 inhibitor-based therapies:
 - ORR 45.9%; median DoR 10.9 months; median PFS 8.3 months.
- The spectrum of *HER2* mutations is consistent with previously evaluated SUMMIT cohorts and with literature-reported prevalence.
- While the rate of grade 3 diarrhea was higher than that observed with single-agent neratinib in SUMMIT, this was manageable through loperamide prophylaxis. No patient discontinued treatment due to diarrhea.
- SUMMIT has recently been amended to evaluate neratinib + fulvestrant + trastuzumab, trastuzumab + fulvestrant and fulvestrant alone (1:1:1 randomization) and continues to enroll patients:
 - Patients who receive single-agent fulvestrant or fulvestrant + trastuzumab are eligible to crossover to triplet therapy upon progression.

Acknowledgements

- The authors would like to thank all patients and their families for participating in the SUMMIT study.
- SUMMIT was sponsored by Puma Biotechnology Inc.
- Puma Biotechnology Inc. funded medical writing/editing assistance for this poster, which was provided by Miller Medical Communications Ltd.

References

1. Bose et al. Cancer Discovery 2013;3:224–37.
2. Razavi et al. Cancer Cell 2018;34:427–38.
3. Nayar et al. Nat Genet 2019;51:207–16.
4. Croessmann et al. Clin Cancer Res 2019;25:277–89.
5. Hyman et al. Nature 2018;554:189–94.
6. Smyth et al. Cavncer Discov 2020;10:198–213.
7. Ma et al. Clin Cancer Res 2017;23:5687–95.

Copyright 2020 Puma Biotechnology

Presented at the San Antonio Breast Cancer Symposium® - December 8–11, 2020.

This presentation is the intellectual property of the author/presenter. Contact them at Email: jhaverik@mskcc.org for permission to reprint and/or distribute.