Neratinib in *HER2*-mutant, recurrent/metastatic cervical cancer: updated findings from the phase 2 SUMMIT basket trial

Claire F. Friedman,[1] Anishka D'Souza,[2] Anna Tinker,[3] Elena Corral,[4] Valentina Gambardella,[5] Jonathan Goldman,[6] Sherene Loi,[7] Michelle E. Melisko,[8] Ana Oaknin,[9] Iben Spanggaard,[10] Ari VanderWalde,[11] Aimee L. Frazier,[12] Bo Zhang,[12] Lisa D. Eli,[12] David B. Solit[1]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]USC Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [3]BC Cancer-Vancouver, Vancouver, British Columbia, Canada; [4]Ramón y Cajal University Hospital, Madrid, Spain; [5]Hospital Clínico Universitario de Valenci, Valencia, Spain; [6]The David Geffen School of Medicine at UCLA, Los Angeles, CA, USA; [7]Peter MacCallum Cancer Centre, Melbourne, Victoria, Australia; [8]UCSF Early Phase Investigational Therapeutics, University of California San Francisco, San Francisco, CA, USA; [9]Vall d'Hebron University Hospital, Vall d'Hebron Institute of Oncology (VHIO), Barcelona, Spain; [10]University Hospital, Rigshospitalet, Denmark; [11]West Cancer Center and Research Institute, Germantown, TN, USA; [12]Puma Biotechnology Inc, Los Angeles, CA, USA

#559P

Background

- Standard first-line treatment for recurrent or metastatic cervical cancer is platinum-based chemotherapy + bevacizumab, with pembrolizumab + platinum-based chemotherapy ± bevacizumab recommended for patients with programmed death ligand-1 (PD-L1)-positive tumors:[1]
 - Second-line and subsequent therapies include a range of active agents;[1] however, the highest expected response rate (tisotumab vedotin) has not exceeded 24%,[2,3] and there is a recognized need for new treatment options for patients who have progressed on or after platinum-based therapy.
 - Currently, no targeted therapies have been developed for metastatic cervical cancer.
- Somatic *HER2* mutations are oncogenic drivers in a range of solid tumors:
 - *HER2* mutations are present in ~5% of cervical cancers, and are enriched in adenocarcinoma compared with squamous cell carcinomas.[4-6]
 - Given the clinical utility of HER2-targeted therapies in patients with breast and other cancers, patients with cervical cancers harboring *HER2* mutations may potentially benefit from HER2-directed therapy.
- Neratinib is an irreversible pan-HER tyrosine kinase inhibitor that is approved for HER2-positive (amplified/overexpressed) early and metastatic breast cancer.[7,8]
 - Neratinib displays potent inhibition of cell proliferation and tumor growth in multiple preclinical models, including *HER2*-mutant uterine cervical cancer cell lines and xenografts.[9]
 - Neratinib has demonstrated promising single-agent activity in multiple *HER2*-mutant cancers, including cervical cancer, within the context of the SUMMIT basket trial (Clinicaltrials.gov: NCT01953926).[10,11]
 - We describe final findings from the *HER2*-mutant cervical cancer cohort of the SUMMIT trial.

Methods

Study design

- SUMMIT is an international, open-label, multi-cohort, multi-tumor, phase 2 basket trial (**Figure 1**).
- Patients with persistent, recurrent or metastatic cervical cancer and a *HER2* mutation documented by institutional testing at a CLIA/CAP- (or regionally-equivalent) certified laboratory were eligible for the cervical cancer cohort.

Figure 1. **SUMMIT study design: Neratinib monotherapy cohorts**

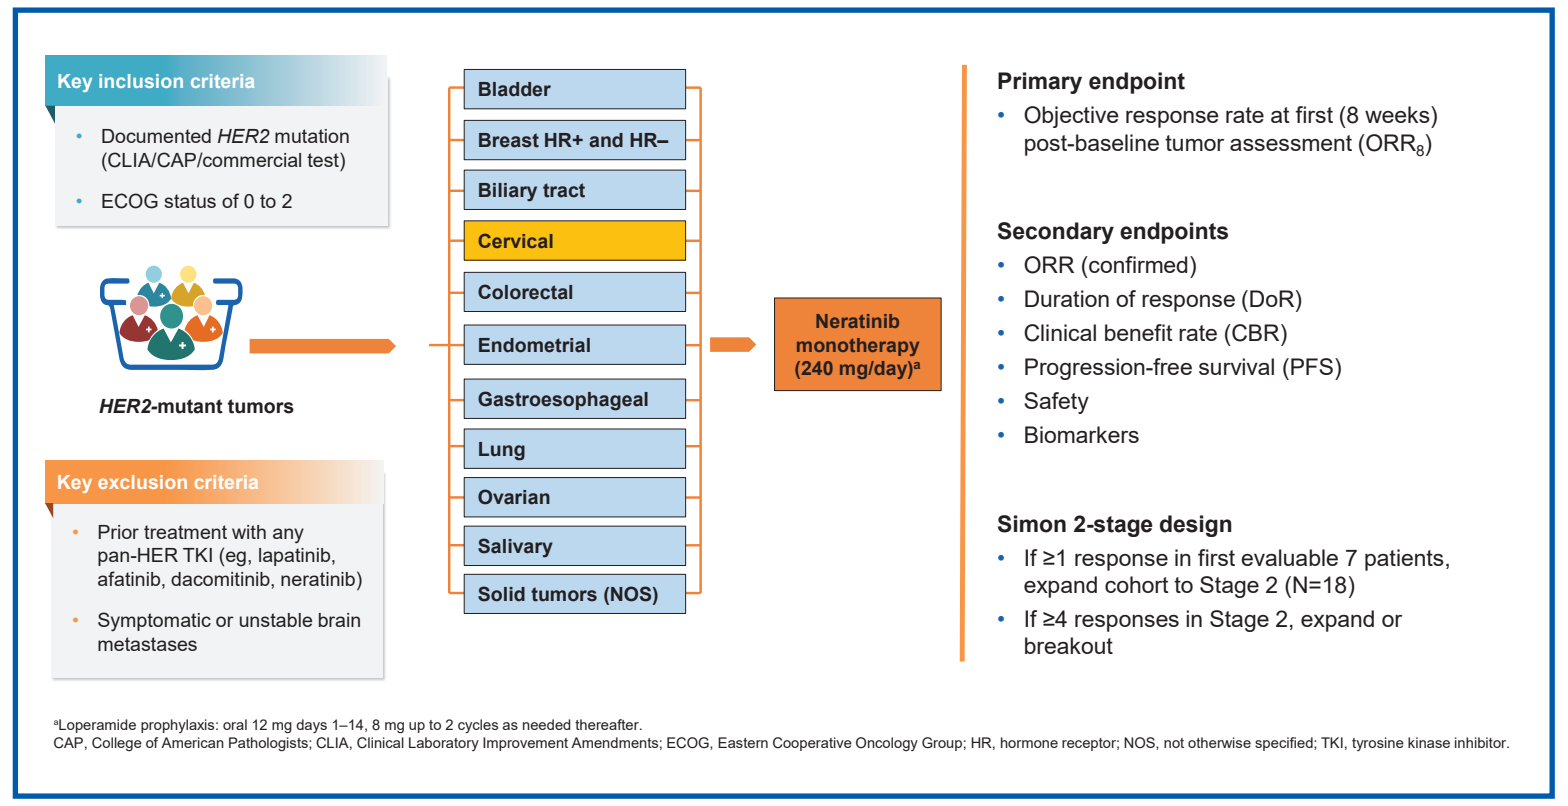
Key inclusion criteria
Documented HER2 mutation (CLIA/CAP/commercial test)
ECOG status of 0 to 2
HER2-mutant tumors
Key exclusion criteria
Prior treatment with any pan-HER TKI (eg, lapatinib, afatinib, dacomitinib, neratinib)
Symptomatic or unstable brain metastases
Bladder
Breast HR+ and HR–
Biliary tract
Cervical
Colorectal
Endometrial
Gastroesophageal
Lung
Ovarian
Salivary
Solid tumors (NOS)
Neratinib monotherapy (240 mg/day)a
Primary endpoint
Objective response rate at first (8 weeks) post-baseline tumor assessment (ORR8)
Secondary endpoints
ORR (confirmed)
Duration of response (DoR)
Clinical benefit rate (CBR)
Progression-free survival (PFS)
Safety
Biomarkers
Simon 2-stage design
If ≥1 response in first evaluable 7 patients, expand cohort to Stage 2 (N=18)
If ≥4 responses in Stage 2, expand or breakout
aLoperamide prophylaxis: oral 12 mg days 1–14, 8 mg up to 2 cycles as needed thereafter.
CAP, College of American Pathologists; CLIA, Clinical Laboratory Improvement Amendments; ECOG, Eastern Cooperative Oncology Group; HR, hormone receptor; NOS, not otherwise specified; TKI, tyrosine kinase inhibitor.

Assessments

- Tumor response was assessed every 8 weeks by investigators using Response Evaluation Criteria in Solid Tumors (RECIST) version 1.1 and/or Positron-Emission Tomography Response Criteria (PERCIST).
- Adverse events were monitored until day 28 after discontinuation of study treatment and classified according to Common Terminology Criteria for Adverse Events version 4.0.

Statistical analyses

- Efficacy and safety outcomes were summarized in the safety analysis set, which included all patients who received at least one dose of neratinib.
- Kaplan–Meier method was used to estimate time-to-event endpoints.
- Clopper–Pearson method was used to calculate 95% confidence intervals for response rates.
- Data cut-off: July 15, 2022.

Results

Patients

- As of July 15, 2022, 22 patients with recurrent or metastatic *HER2*-mutant cervical cancer were enrolled (**Table 1**).
- Median age was 55 (range, 29–74) years, and most patients had adenocarcinomas (82%).
- Prior systemic treatments included platinum-based chemotherapy (100%), bevacizumab (73%), and pembrolizumab (18%).
- Enrolled patients had one (n=18) or two (n=4) *HER2* mutations:
 - Single *HER2* mutations: S310F/Y (n=10); R678Q (n=2); D769H/N (n=2); other (n=4).
 - Double *HER2* mutations included S310F/Y in 3 cases.
- Median duration of neratinib therapy was 3.7 (range, 0.5–67.5) months.

Table 1. **Baseline characteristics**

	HER2-mutant cervical cancer cohort (N=22)
Median age (range), years	55 (29–74)
Race, n (%)	
White	15 (68.2)
Asian	1 (4.5)
Black or African American	1 (4.5)
Unknown or other	5 (22.7)
ECOG performance status, n (%)	
0	7 (31.8)
1	15 (68.2)
FIGO stage at diagnosis (2018 criteria), n (%)	
Stage I	7 (31.8)
Stage II	5 (22.7)
Stage III	2 (9.1)
Stage IV	6 (27.3)
Missing	2 (9.1)
Histology, n (%)	
Adenocarcinoma	18 (81.8)
Squamous	4 (18.2)
HPV status, n (%)	
Positive	13 (59.1)
Negative	2 (9.1)
Unknown/missing	7 (31.8)
Prior radiotherapy, n (%)	17 (77.3)
Prior surgery, n (%)	14 (63.6)
Prior systemic treatments, n (%)	
Platinum-based chemotherapy	22 (100.0)
Bevacizumab	16 (72.7)
Pembrolizumab	4 (18.2)

FIGO, Federation of Gynecology and Obstetrics; HPV, human papillomavirus.

- The spectrum of *HER2* mutations in the SUMMIT cervical cohort is consistent with that reported in public datasets including Project GENIE (**Figure 2**).
- The majority of *HER2* mutations observed in cervical cancer are at position S310 in the extracellular domain.

Figure 2. **Spectrum of *HER2* mutations**

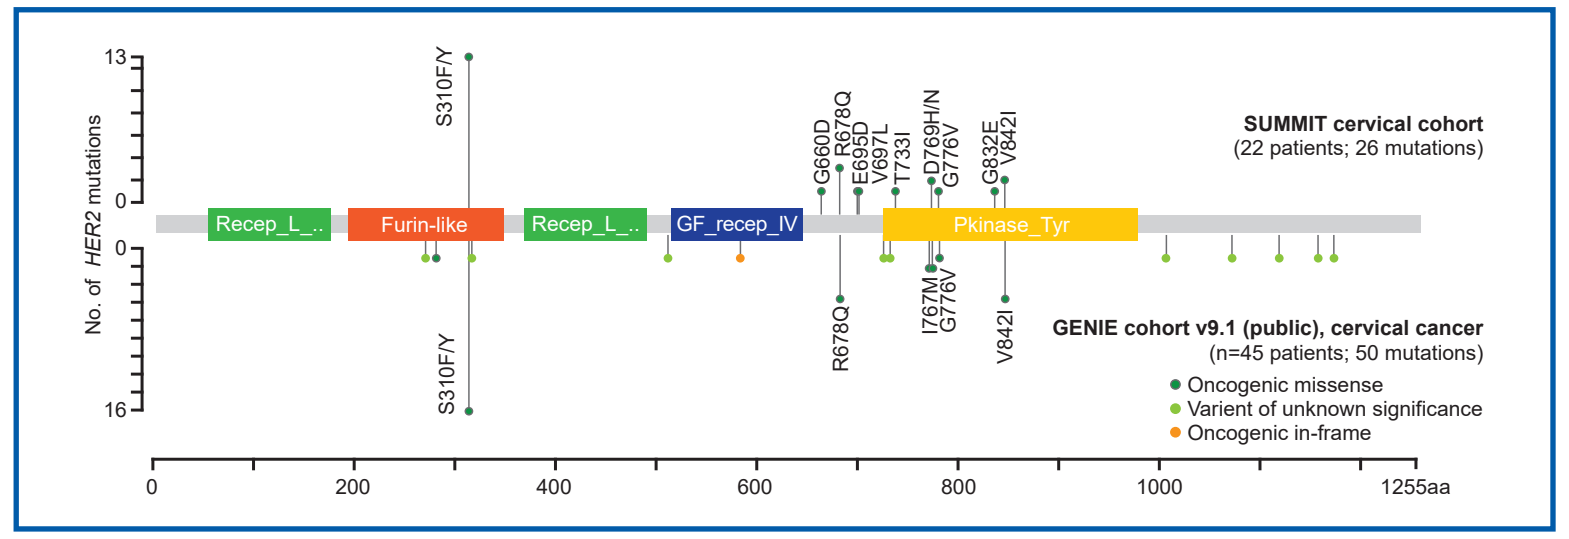
No. of HER2 mutations
13
0
0
16
S310F/Y
G660D
R678Q
E695D
V697L
T733I
D769H/N
G776V
G832E
V842I
R678Q
I767M
G776V
V842I
S310F/Y
Recep_L_..
Furin-like
Recep_L_..
GF_recep_IV
Pkinase_Tyr
SUMMIT cervical cohort
(22 patients; 26 mutations)
GENIE cohort v9.1 (public), cervical cancer
(n=45 patients; 50 mutations)
Oncogenic missense
Varient of unknown significance
Oncogenic in-frame
0
200
400
600
800
1000
1255aa

Efficacy

- Key findings with neratinib monotherapy (**Table 2**):
 - Confirmed ORR: 18.2%.
 - Median DoR: 7.6 months.
 - CBR: 45.5%.
 - Median PFS: 5.1 months.
- Five patients had sustained clinical benefit lasting for 12 months or longer (S310F, n=4; D769N, n=1) (**Figure 3**).
- Prior use of bevacizumab did not appear to influence the efficacy of neratinib (**Figure 4**).
- 55.6% (10/18) of patients with cervical adenocarcinoma experienced clinical benefit, while none (0/4) of the patients with squamous cell carcinoma did so (**Table 2**).

Table 2. **Efficacy**

	Overall	Histology		*HER2* mutation				
Endpoint	*HER2*-mutant cervical cancer cohort (N=22)	Adenocarcinoma (n=18)	Squamous cell carcinoma (n=4)	S310F or S310Y (single mutation) (n=10)	S310F or S310Y (double mutation) (n=3)	D769H or D769N (n=2)	Other kinase domain mutation[a] (n=2)	Transmembrane domain mutation[b] (n=5)
Objective response, n (%)	4 (18.2)	4 (22.2)	0	3 (30.0)	1 (33.3)	0	0	0
CR*	1 (4.5)	1 (5.6)	0	1 (10.0)	0	0	0	0
PR	3 (13.6)	3 (16.7)	0	2 (20.0)	1 (33.3)	0	0	0
ORR, % (95% CI)	18.2 (5.2–40.3)	22.2 (6.4–47.6)	0 (0–60.2)	30.0 (6.7–65.2)	33.3 (0.8–90.6)	0 (0–84.2)	0 (0–84.2)	0 (0–52.2)
Median DoR, months (95% CI)	7.6 (5.6–12.3)	7.6 (5.6–12.3)	0	9.3 (5.6–12.3)	5.9 (NE–NE)	0	0	0
Clinical benefit, n (%)	10 (45.5)	10 (55.6)	0	4 (40.0)	2 (66.7)	2 (100.0)	0	2 (40.0)
CR	1 (4.5)	1 (5.6)	0	1 (10.0)	0	0	0	0
PR	3 (13.6)	3 (16.7)	0	2 (20.0)	1 (33.3)	0	0	0
SD (≥16 weeks)	6 (27.3)	6 (33.3)	0	1 (10.0)	1 (33.3)	2 (100.0)	0	2 (40.0)
CBR, % (95% CI)	45.5 (24.4–67.8)	55.6 (30.8–78.5)	0 (0–60.2)	40.0 (12.2–73.8)	66.7 (9.4–99.2)	100 (15.8–100)	0 (0–84.2)	40.0 (5.3–85.3)
Median PFS, months (95% CI)	5.1 (1.7–7.2)	5.5 (3.7–10.9)	1.7 (0.7–3.5)	3.7 (0.5–14.1)	6.5 (1.7–7.7)	11.9 (5.5–18.3)	NE (1.7–NE)	5.1 (1.7–7.2)

Cut-off date: July 15, 2022. *Complete metabolic response (PERCIST evaluated; no RECIST-evaluable lesion); [a]G776V, G823E. [b]R678Q, V697L, E695D.
CBR, clinical benefit rate; CI, confidence interval; CR, complete response; DoR, duration of response; NE, not estimable; ORR, objective response rate; PFS, progression-free survival; PR, partial response; SD, stable disease.

Figure 3. **Individual response by duration of treatment**

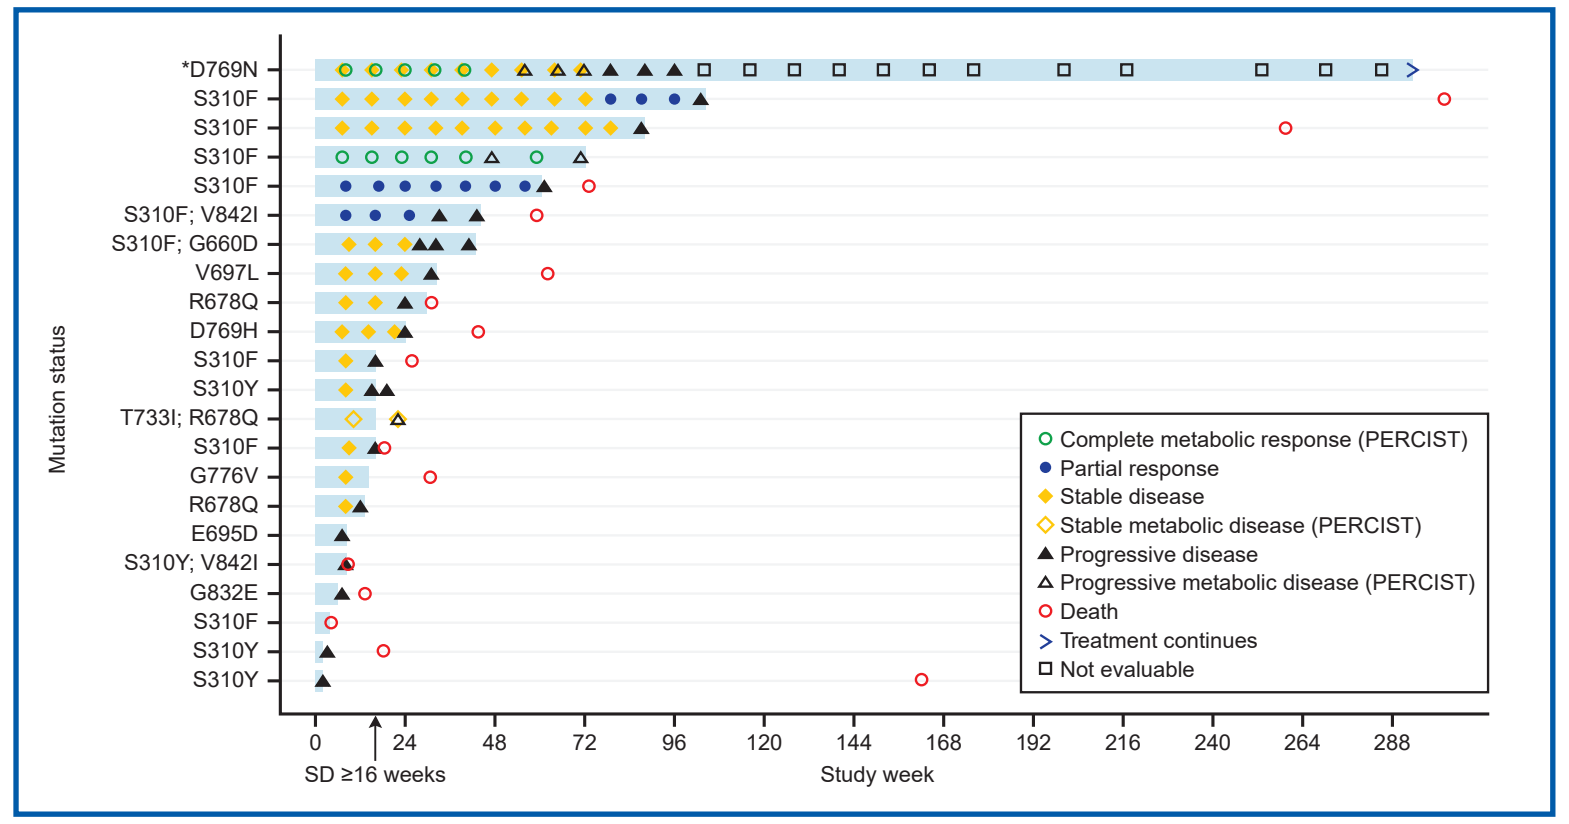
Mutation status
*D769N
S310F
S310F
S310F
S310F
S310F; V842I
S310F; G660D
V697L
R678Q
D769H
S310F
S310Y
T733I; R678Q
S310F
G776V
R678Q
E695D
S310Y; V842I
G832E
S310F
S310Y
S310Y
Complete metabolic response (PERCIST)
Partial response
Stable disease
Stable metabolic disease (PERCIST)
Progressive disease
Progressive metabolic disease (PERCIST)
Death
Treatment continues
Not evaluable
0
24
48
72
96
120
144
168
192
216
240
264
288
SD ≥16 weeks
Study week

Cut-off date: July 15, 2022. *Note: Extreme responder had surgical resection at week 96 and so was not evaluable.

- The majority of SUMMIT cervical cancer patients had tumors that harbored S310F/Y mutation (45.5%, 10/22 with a single S310F/Y mutation plus 13.6%, 3/22 with S310F/Y as one of two *HER2* mutations).
- Confirmed objective response was observed in 30% (3/10) patients whose tumors harbored a single S310F/Y mutation, and 33.3% (1/3) of those with S310 plus one additional *HER2* mutation (**Table 2**).
- Long-term stable disease was observed across multiple *HER2* mutation types (**Table 2**).

Figure 4. **Individual best change in target lesion from baseline (RECIST-evaluable)**

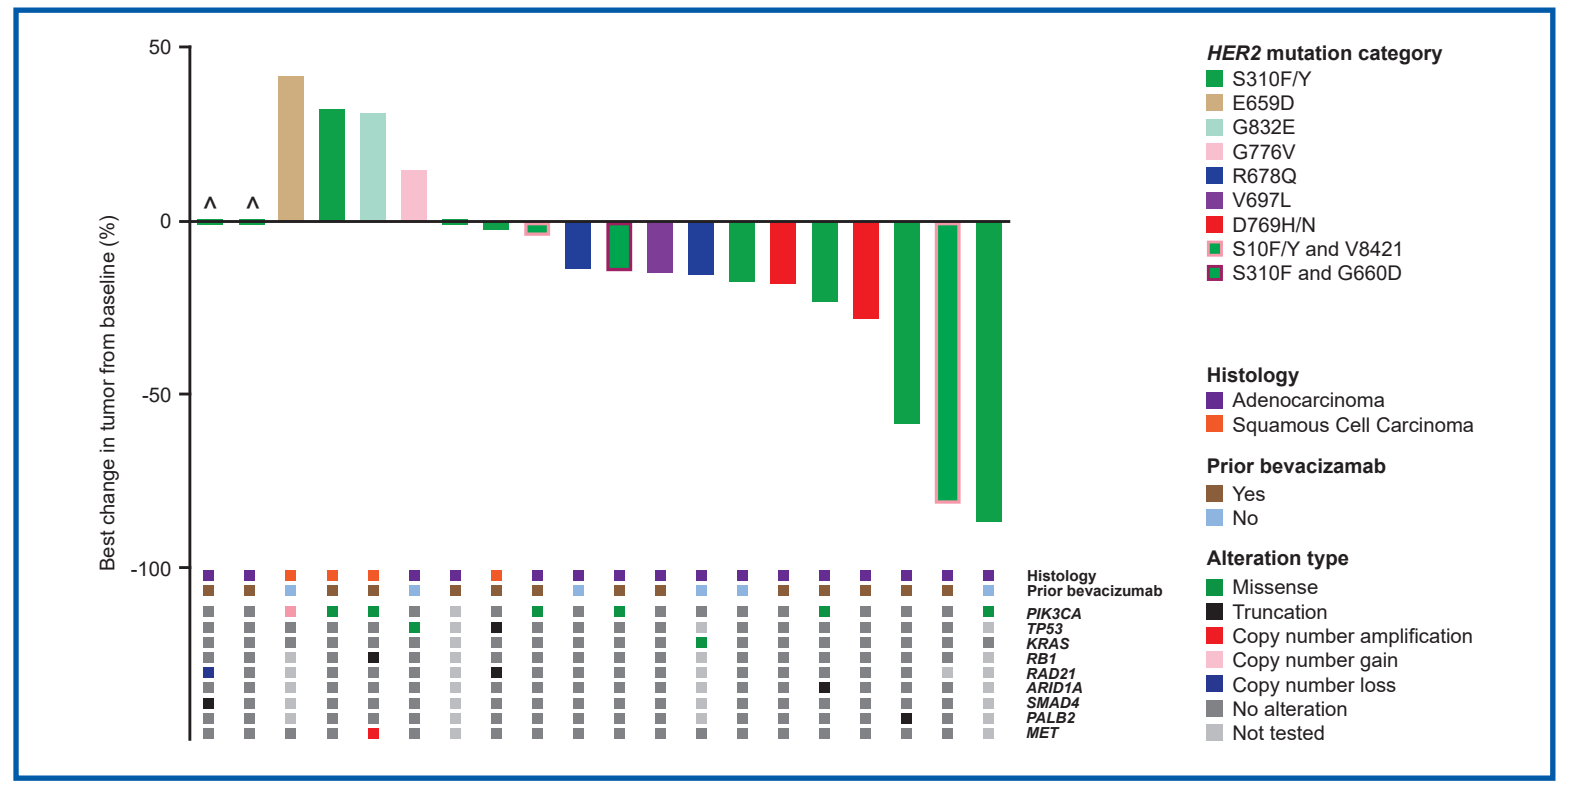
Best change in tumor from baseline (%)
50
0
-50
-100
HER2 mutation category
S310F/Y
E659D
G832E
G776V
R678Q
V697L
D769H/N
S10F/Y and V8421
S310F and G660D
Histology
Adenocarcinoma
Squamous Cell Carcinoma
Prior bevacizumab
Yes
No
Alteration type
Missense
Truncation
Copy number amplification
Copy number gain
Copy number loss
No alteration
Not tested
Histology
Prior bevacizumab
PIK3CA
TP53
KRAS
RB1
RAD21
ARID1A
SMAD4
PALB2
MET

Cut-off date: July 15, 2022. RECIST-evaluable patients at baseline (n=20); 2 other patients not shown were PERCIST evaluable only. Co-mutation data based on local/enrollment assays.

Safety

- Safety profile of neratinib in patients with *HER2*-mutant cervical cancer was consistent with other reports of neratinib monotherapy (Table 3).
- Diarrhea was common but manageable with antidiarrheal prophylaxis.
- No diarrhea events led to treatment discontinuation or dose reductions.

Table 3. **Safety**

Adverse event, n (%)	All adverse events (N=22)		Treatment-related adverse events (N=22)	
	All grades	Grade ≥3[a]	All grades	Grade ≥3
Patients with at least 1 adverse event	22 (100)	10 (45.5)	19 (84.6)	5 (22.7)
Diarrhea	20 (90.9)	5 (22.7)[b]	18 (81.8)	5 (22.7)
Constipation	12 (54.5)	0 (0)	2 (9.1)	0 (0)
Nausea	12 (54.5)	0 (0)	9 (40.9)	0 (0)
Decreased appetite	9 (40.9)	0 (0)	4 (18.2)	0 (0)
Vomiting	9 (40.9)	0 (0)	5 (22.7)	0 (0)
Abdominal pain	8 (36.4)	2 (9.1)	4 (18.2)	1 (4.5)
Dyspepsia	5 (22.7)	0 (0)	2 (9.1)	0 (0)
Dyspnea	5 (22.7)	1 (4.5)	0 (0)	0 (0)
Headache	5 (22.7)	0 (0)	1 (4.5)	0 (0)
Asthenia	4 (18.2)	1 (4.5)	1 (4.5)	0 (0)
Fatigue	4 (18.2)	0 (0)	1 (4.5)	0 (0)
Pain	4 (18.2)	0 (0)	0 (0)	0 (0)
Malaise	4 (18.2)	0 (0)	1 (4.5)	0 (0)
Dysgeusia	4 (18.2)	0 (0)	4 (18.2)	0 (0)
Dry skin	4 (18.2)	0 (0)	2 (9.1)	0 (0)

Cut-off date: July 15, 2022. Table shows events reported in ≥4 patients.
[a]There were no grade 4 adverse events and two grade 5 (fatal) adverse events: dyspnea (n=1) and embolism (n=1). Neither grade 5 event was reported as related to neratinib treatment.
[b]None of the diarrhea events resulted in dose reductions or discontinuation of neratinib in the cervical cancer cohort.

Conclusions

- Neratinib led to durable responses and disease control in patients with metastatic or recurrent *HER2*-mutant cervical cancer.
- The predominance of S310F/Y in the SUMMIT cervical cancer cohort was consistent with the genomic spectrum of *HER2* mutations reported in public datasets for *HER2*-mutant cervical cancer, including Project GENIE:
 - All patients with a confirmed response had tumors harboring an S310F/Y mutation.
- Patients with *HER2*-mutant cervical adenocarcinoma may respond better than those with squamous cell carcinoma, although this is limited by small numbers and may reflect the fact that *HER2* mutations are enriched in adenocarcinoma.
- Neratinib safety profile was consistent with previous reports in metastatic HER2-positive or *HER2*-mutant tumors.
- Diarrhea was not a treatment-limiting toxicity with antidiarrheal prophylaxis.
- These encouraging results support further investigation of neratinib in patients with persistent, metastatic or recurrent *HER2*-mutant cervical cancer following platinum failure.

Acknowledgements

- Puma Biotechnology Inc. provided funding support for SUMMIT, as well as editorial support by Miller Medical Communications.

References

1. NCCN Clinical Practice Guidelines in Oncology. Cervical Cancer, Version 1, 2022.
2. Coleman RL, et al. Lancet Oncol 2021;22:609–19.
3. Hong DS, et al. Clin Cancer Res 2020;26:1220–28.
4. Schram A, et al. AACR 2017, abstract LB-103.
5. Ojesina AI, et al. Nature 2014;506:371–375.
6. Xiang L, et al. Gynecol Oncol 2018;148:311–316.
7. Chan A, et al. Lancet Oncol 2016;17:367–377.
8. Saura C, et al. J Clin Oncol 2020;38:3138–3149.
9. Lopez S, et al. SGO Meeting 2015, poster 356.
10. Hyman DM, et al. Nature 2018;554:189–194.
11. Oaknin A, et al. Gynecol Oncol 2020;159:150–156.

COI disclosures: Claire Friedman

Financial

- Seagen: consultant.
- BMS: consultant.

Non-financial

- Merck: LYNK-002 Scientific Advisor Board (compensation waived).
- Genentech: MyPathway Scientific Advisor Board.

Copyright 2022 Puma Biotechnology

Presented at the ESMO Congress, 9–13 September 2022, in Paris, France

This presentation is the intellectual property of the author/presenter. Contact them at Email: friedmac@mskcc.org for permission to reprint and/or distribute